HIV

Magellan Rx MANAGEMENT

Real World Health Plan Data Analysis: Key Trends in Medication Adherence and Overall Costs in Patients with HIV

Anne Kangethe[a], Michael Polson[a], Todd C. Lord[a], Themmi Evangelatos[a], Alan Oglesby[b]

[a]Magellan Rx Management • Scottsdale, AZ
[b]ViiV Healthcare • Research Triangle Park, NC

AMCP Nexus 2017 | Dallas, TX

Background

- The CDC estimates that more than 1.2 million people in the US are living with human immunodeficiency virus (HIV)
- There are approximately 56,000 new HIV infections and over 8,000 Acquired Immune Deficiency Syndrome (AIDS) deaths each year in the US[1]
- Since the mid-1990's, highly active antiretroviral therapy (ART) regimens have dramatically improved outcomes for patients with HIV resulting in an 8-fold decrease in mortality rates through 2015[2]
- However, optimum care for these individuals requires high adherence to effective ART[3]
- The introduction of single tablet regimens (STR) has resulted in improved adherence and decreased healthcare costs and hospitalizations for patients with HIV compared to existing multiple tablet regimens (MTR) in real world settings[4-6]
- However, the relationship between adherence and healthcare costs may be more complicated than previously thought[7]
- This study aims to determine the key trends in adherence and costs of care for patients with HIV

Objective

- The purpose of this project was to conduct a retrospective evaluation of health plan medical and pharmacy data to assess overall ART adherence trends in patients with HIV
- The impact of medication adherence on healthcare costs were described in the context of medication formulation and follow-up time

Methods

Data Source & Study Time Period

- This retrospective study analyzed Magellan's medical and pharmacy claims data from 1/1/2007-6/30/2016 for commercially-insured patients continuously enrolled for six months prior to the index date and at least 15 months after the index date
 - The index date was the first medical claim with a diagnosis of HIV or for an HIV ART during the index period (July 1, 2007 - June 30, 2014)
- Baseline metrics were measured in the six month period prior to the index date and patients were followed for at least 15 months post-index

Inclusion Criteria

- A diagnosis of human immunodeficiency virus (HIV) ICD-9 042 or ICD-10 CM B20 or asymptomatic HIV infection ICD-9 V08 or ICD-10 CM Z21 in the index period or at least one pharmacy claim for HIV ART at index
- At least two pharmacy claims for HIV antiretroviral medication during the follow-up period
- Age ≥ 18 years of age on the index date
- Continuous eligibility for both medical and pharmacy coverage six months prior to the index date through at least 15 months after the index date

Cohort Assignment

- Each patient was categorized based on ART regimen pill burden: STR or MTR
- Patients were further divided into two cohorts based on length of available follow-up data: < 3 years (cohort 1) or ≥ 3 years (cohort 2)

Descriptive Analysis

- Descriptive statistics were generated to describe baseline continuous (mean, median, standard deviation) and categorical variables (count and percentage)
- Adherence was measured continuously using proportion of days covered [PDC]. High adherence was defined as ≥ 95% PDC
 - PDC assessed the available days' supply of a dispensed medication from pharmacy claims data across the follow-up time for each individual patient[8]

Calculation of Costs

- Cost information, across all encounters regardless of reason, was based on the claim allowed amounts for both medical and pharmacy data
 - Costs were segregated by site of care and classified as medical costs, pharmacy costs and aggregated as total costs
- Costs were reported as annual averages
 - Multivariate adjustments that control for baseline characteristic confounders was analyzed

Results

- A total of 1,698 patients met inclusion criteria; 686 and 1,012 had < 3 and ≥ 3 years of follow-up data respectively (mean 2.1 vs 4.7 years) (Table 1)
- STR patients represented 47% and 37% of the treated subjects in cohorts 1 and 2, respectively
- Mean overall adherence was significantly higher in both cohorts among the STR patients compared to the MTR patients (cohort 1 with 53% vs 39%; cohort 2 with 61% vs 45%, p<0.0001). Similarly, a greater proportion of STR patients in both cohorts achieved high adherence compared to MTR patients (Table 2)
- Not surprisingly, non-adherent patients in both cohorts had numerically lower pharmacy costs as these patients , by definition, filled fewer prescriptions. This difference was significant for cohort 2 (Tables 3 & 4)
- In both cohorts, non-adherent patients had numerically higher mean annual medical costs, but this was significant for cohort 2 only (cohort 1 with $10,572 vs $9,474; p=0.819 and cohort 2 with $8,224 vs $3,097; p<0.001) (Tables 3 & 4)
- However, there was significant savings in mean annual inpatient costs between the adherent and non-adherent patients in both cohorts (cohort 1 with -$2,525; p<0.001 and cohort 2 with -$815; p<0.001) (Tables 3 & 4)

Table 1: Baseline Demographics

	< 3 year data				≥ 3 year data			
	All n=686	STR n=322	MTR n=364	P-value	All n=1012	STR n=377	MTR n=635	P-value
Age								
Continuous	45.10 (11.21) [47.00]	42.52 (11.81) [42.79]	47.38 (10.13) [47.81]	< 0.0001	47.49 (10.12) [48.10]	45.15 (10.99) [46.41]	48.88 (9.30) [49.33]	< 0.0001
18 - 29	83 (12.1%)	59 (18.3%)	24 (6.6%)	< 0.0001	67 (6.6%)	41 (10.9%)	26 (4.1%)	< 0.0001
30 - 39	114 (16.6%)	67 (20.8%)	47 (12.9%)		141 (13.9%)	69 (18.3%)	72 (11.3%)	
40 - 49	230 (33.5%)	100 (31.1%)	130 (35.7%)		367 (36.3%)	135 (35.8%)	232 (36.5%)	
50 - 59	199 (29.0%)	74 (23.0%)	125 (34.3%)		337 (33.3%)	98 (26.0%)	239 (37.6%)	
60 - 69	57 (8.3%)	21 (6.5%)	36 (9.9%)		94 (9.3%)	32 (8.5%)	62 (9.8%)	
70 - 79	3 (0.4%)	1 (0.3%)	2 (0.5%)		6 (0.6%)	2 (0.5%)	4 (0.6%)	
Gender								
F	124 (18.1%)	37 (11.5%)	87 (23.9%)	< 0.0001	225 (22.2%)	72 (19.1%)	153 (24.1%)	0.072077
M	562 (81.9%)	285 (88.5%)	277 (76.1%)		787 (77.8%)	305 (80.9%)	482 (75.9%)	
Pre-index Deyo-Charlson Comorbidity Index (HIV diagnosis not included)								
0	467 (68.1%)	221 (68.6%)	246 (67.6%)	0.172384	761 (75.2%)	291 (77.2%)	470 (74.0%)	0.119283
1	116 (16.9%)	60 (18.6%)	56 (15.4%)		150 (14.8%)	58 (15.4%)	92 (14.5%)	
2	62 (9.0%)	28 (8.7%)	34 (9.3%)		58 (5.7%)	19 (5.0%)	39 (6.1%)	
3+	41 (6.0%)	13 (4.0%)	28 (7.7%)		43 (4.2%)	9 (2.4%)	34 (5.4%)	
Continuous	0.60 (1.20) [0.00]	0.52 (0.97) [0.00]	0.68 (1.37) [0.00]	0.0699	0.43 (0.99) [0.00]	0.35 (0.81) [0.00]	0.48 (1.08) [0.00]	0.0247
3rd Agent ART Class								
NNRTI*	371 (54.1%)	249 (77.3%)	122 (33.5%)	< 0.0001	636 (92.7%)	354 (93.9%)	282 (44.4%)	< 0.0001
PI*	229 (33.4%)	0	229 (62.9%)	< 0.0001	405 (40.0%)	0	405 (63.8%)	< 0.0001
INSTI*	252 (36.7%)	94 (29.2%)	158 (43.4%)	0.000135	304 (44.3%	46 (12.2%)	258 (40.6%)	< 0.0001

Continuous data: mean (standard deviation) [median]
Discrete data: count (percent)
*NNRTI=Non-Nucleoside Reverse Transcriptase Inhibitors; PI=Protease Inhibitors; INSTI=Integrase Strand Transfer Inhibitors

Table 2: Overall Adherence (All Quarters)

Patients with < 3 year data N = 686				
Cohort	**n**	**Overall PDC**	**Standard Deviation**	**P-Value**
MTR	364	0.3941	0.2576	< 0.0001
STR	322	0.5346	0.2804	
Adherence	**n**		**Patient Proportion**	
PDC < .95	620		MTR = 94% STR = 86%	
PDC ≥ .95	66		MTR = 6% STR = 14%	

Patients with ≥ 3 year data N = 1012				
Cohort	**n**	**Overall PDC**	**Standard Deviation**	**P-Value**
MTR	635	0.4533	0.2816	< 0.0001
STR	377	0.6146	0.3053	
Adherence	**n**		**Patient Proportion**	
PDC < .95	906		MTR = 95% STR = 80%	
PDC ≥ .95	106		MTR = 5% STR = 20%	

Table 3: Annual Costs for Patients with < 3 year data

Costs ($)				
Measure	**ALL n=686**	**PDC < 95% n=620**	**PDC ≥ 95% n=66**	**P-Value**
Inpatient Costs	2,690.22 (15,156.47) [0.00]	2,933.18 (15,908.24) [0.00]	407.93 (2,237.24) [0.00]	0.0003
Outpatient Costs	860.09 (1,316.35) [583.51]	868.06 (1,368.19) [582.81]	785.25 (654.12) [621.26]	0.3971
Lab Costs	1,641.20 (10,740.35) [618.96]	1,711.99 (11,280.52) [731.33]	976.19 (1,832.02) [218.46]	0.1465
Emergency Department Costs	319.30 (906.88) [0.00]	299.49 (739.52) [0.00]	505.34 (1,849.36) [0.00]	0.3731
Other Costs	4,955.72 (20,237.87) [653.65]	4,759.46 (17,643.47) [690.69]	6,799.33 (36,714.75) [349.06]	0.6571
Medical Costs	10,466.59 (32,722.45) [2,773.37]	10,572.24 (32,231.48) [2,909.57]	9,474.08 (37,280.27) [1,584.18]	0.8185
Pharmacy Costs	23,706.14 (19,689.24) [21,018.31]	23,384.61 (20,048.65) [20,762.03]	26,726.52 (15,727.96) [23,245.40]	0.1145
Total Costs	34,172.73 (39,095.92) [26,931.98]	33,956.85 (38,279.62) [26,845.79]	36,200.60 (46,355.93) [28,829.07]	0.7053

Table 4: Annual Costs for Patients with ≥ 3 year data

Costs ($)				
Measure	**ALL n=686**	**PDC < 95% n=620**	**PDC ≥ 95% n=66**	**P-Value**
Inpatient Costs	773.22 (5,774.89) [0.00]	858.58 (6,097.67) [0.00]	43.68 (191.71) [0.00]	< 0.0001
Outpatient Costs	780.83 (2,804.31) [481.17]	798.38 (2,958.98) [475.43]	630.88 (480.24) [532.44]	0.1241
Lab Costs	1,062.12 (4,583.09) [211.83]	1,148.39 (4,829.16) [245.52]	324.81 (793.51) [49.76]	< 0.0001
Emergency Department Costs	246.14 (560.54) [0.00]	249.76 (572.59) [0.00]	215.24 (445.39) [0.00]	0.4663
Other Costs	4,824.42 (28,786.50) [466.19]	5,168.58 (30,199.00) [494.65]	1,882.83 (10,425.19) [329.27]	0.0217
Medical Costs	7,687.06 (31,219.13) [1,912.02	8,224.04 (32,758.17) [1,985.07]	3,097.45 (10,559.29) [1,301.72]	0.8185
Pharmacy Costs	19,629.87 (16,745.96) [17,968.69]	27,018.30 (38,323.72) [20,461.75]	29,869.38 (20,209.18) [26,108.10]	0.1145
Total Costs	27,316.93 (36,849.66) [21,376.33]	27,018.30 (38,323.72) [20,461.75]	29,869.38 (20,209.18) [26,108.10]	0.2244

Conclusion

- Overall, adherence is less than optimal for patients with HIV
- Patients on STR were more adherent than patients on MTR regardless of the length of follow-up
- Not surprisingly, higher medication adherence is associated with numerically higher pharmacy costs
- However, higher adherence was associated with significant medical cost savings as patients are continued to be followed over time
- Better adherence was associated with significant inpatient costs savings regardless of follow-up time
- The relationship between adherence and total medical costs is more nuanced than previously reported depending on the follow-up period
- Results suggest further examination of the time-dependency of improved adherence to improved clinical and cost outcomes

References

[1]Barr D. Filling the gaps in the U.S. HIV Treatment Cascade: developing a community-driven research agenda. http://www.treatmentactiongroup.org/hiv/filling-gaps. Accessed July 29, 2016.

[2]CDC. *Mortality Slide Series through 2014*, https://www.cdc.gov/hiv/pdf/library/slidesets/cdc-hiv-mortality.pdf. Accessed April 25, 2017.

[3]Gardner EM, McLees MP, Steiner JF, Del Rio C, Burman WJ. The spectrum of engagement in HIV care and its relevance to test-and-treat strategies for prevention of HIV infection. *Clin Infect Dis.* 2011;52(6):793-800. doi:10.1093/cid/ciq243.

[4]Cohen CJ, Meyers JL, Davis KL. Association between daily antiretroviral pill burden and treatment adherence, hospitalisation risk, and other healthcare utilization and costs in a US Medicaid population with HIV. *BMJ Open.* 2013;3(8):e003028.

[5]Sax PE, Meyers JL, Mugavero M, Davis KL. Adherence to antiretroviral treatment and correlation with risk of hospitalization among commercially insured HIV patients in the United States. *Plos Ones.* 2012;7(2):e31591.

[6]Langness J, Cook PF, Gill J, Boggs R, Netsanet N. Comparison of adherence rates for antiretroviral, blood pressure, or mental health medications for HIV-positive patients at an academic medical center outpatient pharmacy. *J Manag Care Pharm.* 2014;20(8):809-814. doi:10.18553/jmcp.2014.20.8.809.

[7]Pruitt Z, Robst J, Langland-Orban B, Brooks RG. Healthcare costs associated with antiretroviral adherence among medicaid patients. *Appl Health Econ Health Policy.* 2015;13(1):69—80. doi:10.1007/s40258-014-0138-1.

[8]Nau D. Proportion of Days Covered (PDC) as a Preferred Method of Measuring Medication Adherence. Springfield, VA: Pharmacy Quality Alliance; 2012:3. http://ep.yimg.com/ty/cdn/epill/pdcmpr.pdf. Accessed June 6, 2017.

Disclosures

- This research was conducted by Magellan Rx Management, Scottsdale, AZ, with external funding from ViiV Healthcare

ViiV Healthcare